PERSISTENT SKIN SANITIZER (NIMBUDERM™) WITH SUSTAINED MICROBICIDAL PROPERTIES

David Moore[1], William Toreki[1], Roy Carr[1], Bernd Liesenfeld[1], Gregory Schultz[1,2], Christopher Batich[1,2], Paul Dominguez[2], Jillian Vella[1], Gerald Olderman[1], [1]Quick-Med Technologies Inc., [2]University of Florida, *corresponding author*

Poster 99: 20th Annual Symposium on Advanced Wound Care and Wound Healing Society Meeting

Introduction

Quick-Med Technologies, Inc. (QMT) has developed an advanced "leave-on" skin sanitizer formulation, NIMBUDERM™, that provides a unique combination of instant and long-lasting antimicrobial protection. This was achieved by combining the immediate disinfection power of an alcohol-based product, with the long-lasting antimicrobial persistence of an advanced bio-active polymer. Performance at the time of application matches that of common alcohol-based products such as Purell® Hand Sanitizer; however, NIMBUDERM™'s advanced antimicrobial polymer formulation also provides continuous efficacy against microbial contamination for a prolonged period (up to 6 hours in laboratory testing, even after repeated rinsing with water). NIMBUDERM™ provides an invisible antimicrobial polymer shield that persists long after the alcohol has evaporated, offering long-lasting protection against a wide range pathogenic bacteria associated with nosocomial infections. Efficacy has been shown against bacteria including *Staphylococcus aureus*, *Pseudomonas aeruginosa*, as well as antibiotic-resistant organisms such as MRSA and VRE. The advanced NIMBUDERM™ polymer is suitable for incorporation into various formulations and is compatible with common additives such as emollients (to help keep skin soft and hydrated), therapeutic nutrients, and other active ingredients. Antimicrobial skin care products formulated with NIMBUDERM™ technology are the perfect choice for everyday use by both patients and health-care professionals.

Hospital acquired infections are an increasing public health problem

"MRSA has become so common that in many hospitals more than half of all Staph infections tested are drug-resistant."
-USA Today

"The average hospital payment for a Pennsylvania patient who did not have an infection was $8,078, compared with $60,678 for patients who did."
-Pennsylvania Health Care Cost Containment Council, Washington Post

USA Today reported in May 2006 that some hospitals are experiencing an increase of MRSA infections from 33% of all *Staphylococcus aureus* tested in 2000 to 75% in 2006. The CDC reports that nearly 4 per 1000 hospital discharges have acquired an MRSA infection, demonstrating a growing problem within the community from a community health perspective as well as a financial perspective. The Washington Post reports that a Pennsylvania hospital study indicated that 180 hospitals reporting nosocomial infections showed an additional billing of $2.3 billion of which only $614 million was collected from insurance.

Benefits of NIMBUDERM™ Leave-on skin sanitizer
- Provides a bacterial barrier
- Can help prevent transfer of bacteria from nurse to patient, patient to nurse, and patient to patient.
- Can help lower both patient and hospital costs associated with increased nosocomial infections.

Problems with Competitive Skin Sanitizers

- Povidone-Iodine solutions have been proven to show contact dermatitis in as little as 2% Iodine in solution in both allergy sensitive and non-allergy sensitive subjects.
- Povidone-Iodine solutions have been shown to inhibit human fibroblast growth.
- Solutions based solely on alcohol do not persist and can leave the sanitized skin both dry and flaky.
- Hydrogen peroxide solutions have shown a high toxicity index rating against keratinocytes.
- Both Dial Anti-Bacterial and Ivory Liquigel soap formulations have shown a high toxicity index ratings against infant dermal fibroblasts.

(References available upon request)

NIMBUS™ Materials and Methods

The NIMBUS™ family of processes has applications developed that are suitable for a wide variety of substrates, to render the material of choice antimicrobial. The details of the binding are specific to the substrate and application. The antimicrobial agent is in polymeric form, and enables the binding of a second species for release if this is desired.

Thin Film Efficacy Test

The Thin Film Efficacy Test (TFET) was used to determine the bacteriostatic ability of the antimicrobial solution. Growth media plates are used as carriers in which 100 µl of antimicrobial solution is applied to the center of the plate. The coated plates are inoculated with 1000 µl inoculum (titer of 10^6 CFU/ml.) The inoculated plates are allowed to dry and incubated overnight at 37° C, then observed for suppression of bacterial growth. If growth is observed, the plate is considered failing.

ASTM E 1874-97, "Standard Test Method for Evaluation of Antibacterial Washes by Cup Scrub Technique."

Samples were cut from a sheet of pig skin, stratum corneum side up. 500 µl of test product (NIMBUDERM™ or placebo) was applied to each sample (within the designated square) and the sterile scrubbing cup was centered onto the application site to form a cup/skin seal. 250 µl of inoculum were pipetted into the cup, and left for a 5 minute exposure. After exposure, a sterile glass rod was used to scrub the skin within the cup for 30 seconds (Figure 4) and the solution was recovered with a sterile pipette into 0.5 mL of neutralizer (Figure 5). Quantitative analysis was performed by plating and serial dilution using standard techniques.

Thin Film Efficacy Test Results for NIMBUDERM™ Versus Purell®

Skin Sanitizers	*S. aureus*		*E. coli*		MRSA	VRE
	24 Hrs Results	48 Hrs Results	24 Hrs Results	48 Hrs Results	24 Hrs Results	24 Hrs Results
Nimbuderm	**60 Pass/ 0 Fail**	**60 Pass/ 0 Fail**	**60 Pass/ 0 Fail**	**60 Pass/ 0 Fail**	**60 Pass/ 0 Fail**	**60 Pass/ 0 Fail**
Purell®	**0 Pass/ 60 Fail**	**0 Pass/ 60 Fail**	**0 Pass/ 60 Fail**	**0 Pass/ 60 Fail**		

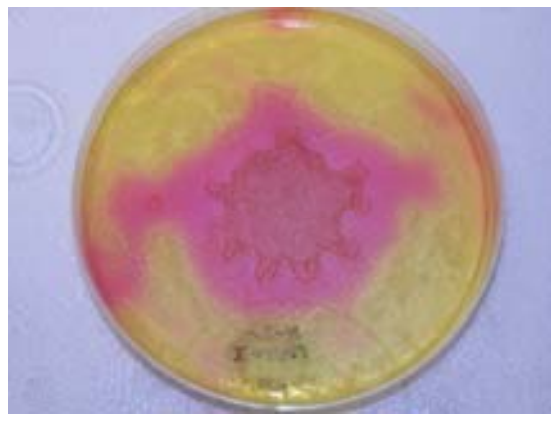

Figure 2 (above). Mannitol Salt Agar (MSA) plate inoculated with *S. aureus* after NIMBUDERM™ application. MSA appears yellow where *S. aureus* metabolizes the mannitol. Pink area represents NIMBUDERM™ applied area in which there are no organisms present.

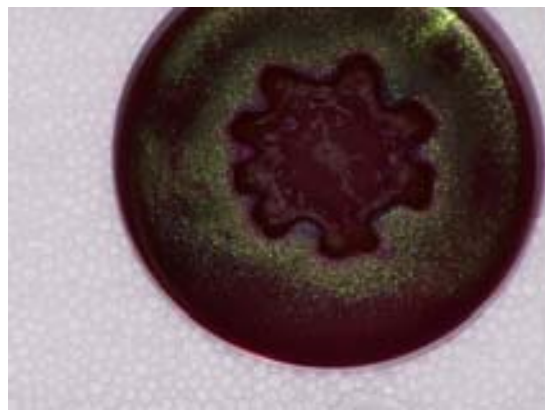

Figure 3 (above). Eosin Methylene Blue (EMB) agar plate inoculated with *E. coli* after NIMBUDERM™ application. EMB appears translucent green where *E. coli* is present.

Bactericidal Persistence Efficacy of NIMBUDERM™ Skin Sanitizer Versus Purell® Instant Hand Sanitizer

Kill levels for:	% killed NIMBUDERM	% killed Purell®
Staphylococcus aureus[1,2] (ATCC #6538)	>99.9999%	<0.0%
Escherichia coli[1,2] (ATCC #15597)	>99.9999%	<0.0%
Pseudomonas aeruginosa[1,2] (ATCC #15442)	>99.9999%	<0.0%
Serratia marcescens[1,2] (ATCC #13880)	>99.9999%	
MRSA[2] (ATCC #BAA-44)	>99.9999%	
Vancomycin Resistant Enterococcus (VRE)[2] (ATCC #700221)	>99.9999%	

Test method used was modified AOAC Use-Dilution Test on [1]glass slide and/or [2]pig skin carriers at a 4 hour exposure - drying time for sanitizer ranged from 1 hour to 3 hours

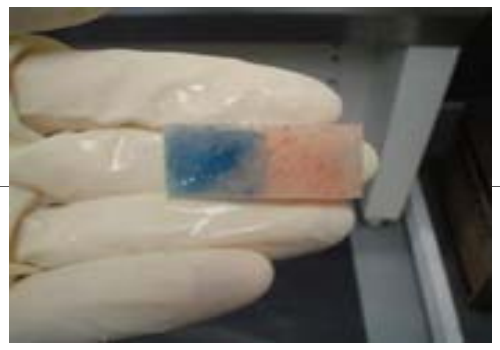

Figure 1 (left). Section of pig skin partially treated with NIMBUDERM™ (left half) after rinsing followed by saturation with Bromothymol blue dye solution. Blue area represents coverage of the treatment due to the electrostatic interactions between the dye and NIMBUDERM™.

Bactericidal Persistence of NIMBUDERM™ Skin using Cup-Scrub Methodology

Kill levels for:	Percent killed
MRSA (ATCC #BAA-44)	>99.998%*
Escherichia coli (ATCC #15597)	>99.997%*
Serratia marcescens (ATCC #13880)	>99.98%*
Serratia marcescens (ATCC #14756)	>99.9998%*
Vancomycin Resistant Enterococcus (VRE) (ATCC #700221)	>99.99999%*

Cup Scrub Results using Human Subjects Demonstrating Time Persistence and Rinsing** Persistence

Drying time of sanitizer on human skin	Percent kill levels for *Serratia marcescens* (ATCC #13880)
T=0	>99.98%*
T=4	>99.99999%*
T=6	>99.99999%*

% kill levels for *S. marcescens* (ATCC #14756)	Efficacy after 0 rinses	Efficacy after 1 rinse
NIMBUDERM™ applied to subject 1	>99.9987%*	>99.9987%*
NIMBUDERM™ applied to subject 2	>99.973%	>99.72%
NIMBUDERM™ applied to subject 3	>99.9993%*	>99.9993%*

***indicates full kill**
**** Rinsing step includes 20X applications (~1 ml each) to sanitizer applied area from standard spray bottle.**

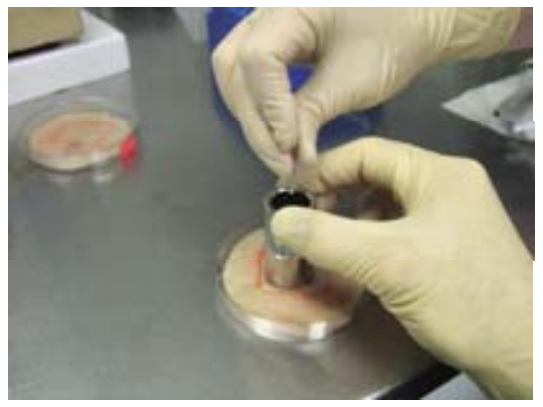

Figure 4 (above). Scrubbing of pig skin treated with NIMBUDERM™.

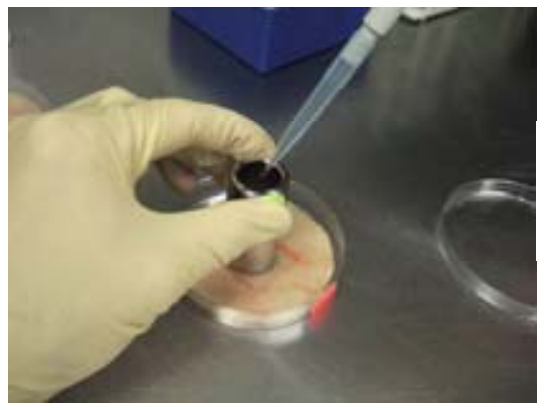

Figure 5 (above). Recovery of organisms after the scrubbing process prior to enumeration.

Safety Testing Data: Data for Related NIMBUS™ Products

*Testing performed on NIMBUS™ treated cotton and rayon materials, by Geneva Test Labs, PO Box 140, Elkhorn, WI

Rabbit eye irritation
- **Results: Non-irritating** Protocol CL 1003

Rabbit Skin Irritation
- **Results: Non-irritating** Protocol CL 1005

Guinea Pig Dermal Sensitization
- **Results: Non-sensitizing** Protocol CL 1015

WWW.QUICKMEDTECH.COM

David Lerner - President (561) 750-4202
Email: dlerner@quickmedtech.com

Roy Carr – Director of Research & Development, Medical Devices (630) 214-9819
Email: rcarr@quickmedtech.com

Gerald M. Olderman - Ph.D. VP, Research & Development (781) 271-9893
Email: jolderman@aol.com